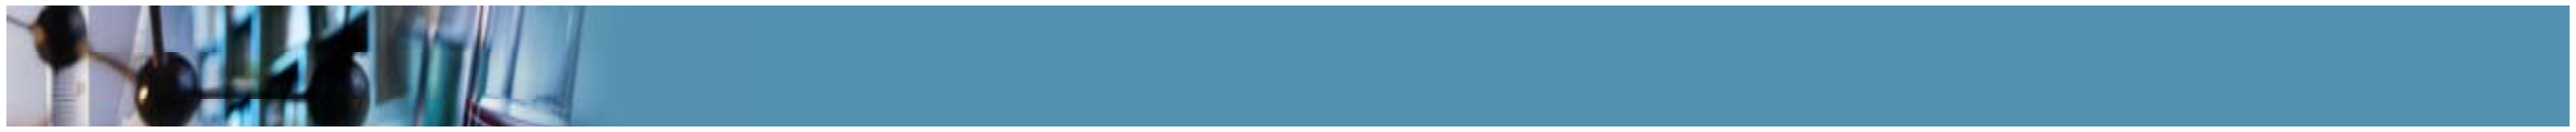

Lead Optimization of Novel Boron-Containing Drug Candidates for the Treatment of Human African Trypanosomiasis

Robert Jacobs[1], Charles Ding[2], Yvonne Freund[2], Kurt Jarnagin[2], Jacob Plattner[2], Cyrus Bacchi[3], Nigel Yarlett[3], Matthew Orr[1], Bakela Nare[1], Cindy Rewerts[1], Daitao Chen[1], Andy Noe[1], Jessica Sligar[1], Matthew Jenks[1], Stephen Wring[1], Robert Don[4].

[1]*SCYNEXIS, Inc., Research Triangle Park, NC, United States,* [2]*Anacor Pharmaceuticals, Inc., Palo Alto, CA, United States,* [3]*Pace University, New York, NY, United States,* [4]*Drugs for Neglected Disease initiative, Geneva, Switzerland.*

Abstract

Human African Trypanosomiasis (HAT) represents a significant public health problem in sub-Saharan Africa affecting hundreds of thousands of individuals. An urgent need exists for the discovery and development of new, safe, and effective drugs to treat HAT, as existing therapies suffer from poor safety profiles, difficult treatment regimens, limited effectiveness, and a high cost of goods. From a collaborative effort between SCYNEXIS, Anacor Pharmaceuticals, Pace University, and DND*i*, we report ongoing lead optimization efforts on a novel class of small molecule boron-containing compounds, exemplified by SCYX-6759. These compounds inhibit *in vitro* growth of *T. brucei* with IC_{50}'s ~100 nM, are not cytotoxic to mammalian cells, and exhibit good physiochemical and pharmacokinetic properties. In a murine model of CNS-stage disease utilizing the TREU667 strain of *T.b.brucei*, treatment with 50 mg/kg of SCYX-6759 BID for 14 days has demonstrated 100% efficacy, resulting in absence of blood parasites for >180 days. Development of a structure-activity relationship (SAR) profile for this chemical series and efforts to improve biological and pharmacokinetic profiles in the CNS-disease model through chemical modifications are reported herein.

Results

Oxaborole 6N-Benzamides

- Active against *T.b.brucei in vitro*, with good PK and physiochemical properties
- Efficacious *in vivo* against both acute (Stage I) and CNS (Stage II) infection in mouse model
- Exemplified by SCYX-6759

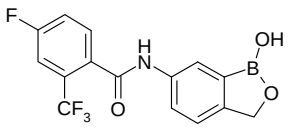

SCYX-6759

T.b.brucei IC_{50} = 180 nM
Cytotoxicity (L929) > 25 μM
LogD = 2.57
Solubility (pH 7.4) > 200 μM
S9 $t_{1/2}$ > 350 min (mouse, rat, human)
P_{app} = 379 nm/s (AQ = 0.02)
Oral Bioavailablity (mouse) = 63%

- Series limited by poor brain disposition and rapid clearance from CNS
- Progressed to lead optimization in mid-2008

Acute *In Vivo* Efficacy (Stage I)

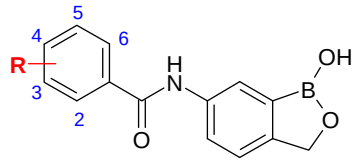

SCYX Id	R	T.b. brucei IC_{50} (nM)	L929 Cytotoxicity IC_{50} (μM)	Animals cured * (10 mg/kg, p.o., b.i.d.)
4424	H	160	> 39.5	NT
7582	2-F	130	> 36.9	1 / 3
5896	2,6-F_2	159	> 34.5	1 / 3
4461	2-CF_3	125	11.8	3 / 3
7620	2-Cl	70	> 34.8	3 / 3
6835	2-Cl, 4-F	147	> 32.7	3 / 3
6752	2-CF_3, 5-F	130	15.5	3 / 3
6759	2-CF_3, 4-F	180	> 29.5	3 / 3 **

- Series demonstrates robust *in vivo* activity in mouse model
- *Cure = No parasitemia detectable for >30 days
- ** Also fully efficacious at 5 mg/kg, p.o., b.i.d. (3/3 cured)

SAR Development

- Aryl group necessary for good activity
- 2-CF_3 or 2-Cl increase potency
- 4-F improves metabolic stability

- 5-F well tolerated
- 5-OCH_3, 7-OCH_3, and 7-CH_3 lead to loss of activity

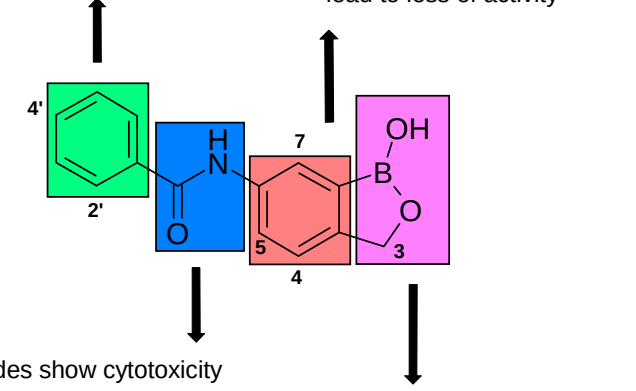

- Sulfonamides show cytotoxicity and poor PK profiles.
- Ureas demonstrate limited permeability
- Amines and carbamates poorly tolerated

- Boron required for activity
- Reduced activity with increasing bulk at 3-position
- **3-Substitution improves PK** with slight loss in potency

Addressing Pharmacokinetic Liabilities

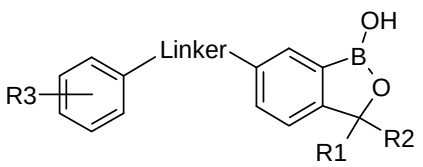

SCYX Id	R_1	R_2	R_3	Linker	T.b.b. IC_{50} (nM)	Cytotox (μM)	MDR1-MDCK Permeability P_{app} (nm/sec)	AQ
6759	H	H	2-CF_3, 4-F	-CONH-	180	> 25	380	0.02
9308	H	CH_3	2-CF_3, 4-F	-CONH-	258	3.4	683	0.03
8258	H	isobutyl	2-CF_3, 4-F	-CONH-	6099	> 25	NT	NT
8240	H	cyclopentyl	2-CF_3, 4-F	-CONH-	7810	> 25	NT	NT
7158	**CH_3**	**CH_3**	**2-CF_3, 4-F**	**-CONH-**	**556**	**> 25**	**415**	**0.03**
2977	Spirocyclopropyl		2-CF_3, 4-F	-CONH-	523	> 25	632	0.08
7265	Spirocyclobutyl		2-CF_3, 4-F	-CONH-	892	> 25	NT	NT
1124	Spirocyclopentyl		2-CF_3, 4-F	-CONH-	13940	> 25	377	-0.02
4435	H	H	4-OCH_3	-SO2NH-	94	6.6	334	0.28
0278	CH_3	CH_3	4-OCH_3	-SO2NH-	6480	>25	NT	NT
6412	H	H	4-Cl	-NHCONH-	238	>25	234	NT
3569	CH_3	CH_3	4-Cl	-NHCONH-	>15000	>25	NT	NT

- SCYX-7158 found to possess the best combination of activity, pharmacokinetics, and physiochemical properties

In Vivo Pharmacokinetics

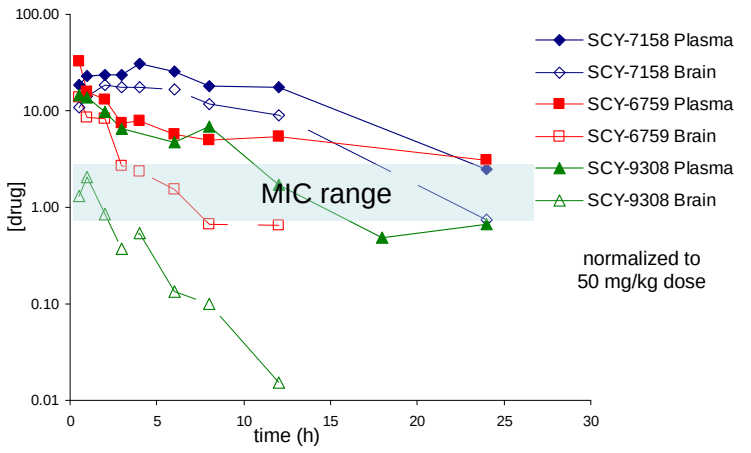
100.00
10.00
1.00
0.10
0.01
[drug]
0
5
10
15
20
25
30
time (h)
MIC range
SCY-7158 Plasma
SCY-7158 Brain
SCY-6759 Plasma
SCY-6759 Brain
SCY-9308 Plasma
SCY-9308 Brain
normalized to
50 mg/kg dose

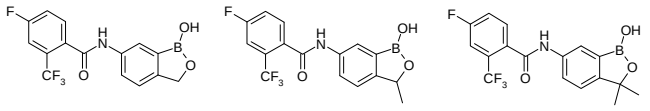

SCYX-6759 — *T.b.brucei* IC_{50} = 180 nM
SCYX-9308 — *T.b.brucei* IC_{50} = 258 nM
SCYX-7158 — *T.b.brucei* IC_{50} = 556 nM

- SCYX-7158 sustains >MIC in CNS for over 24 hours
- Excellent PK and CNS disposition at small cost to potency

CNS (Stage II) Efficacy

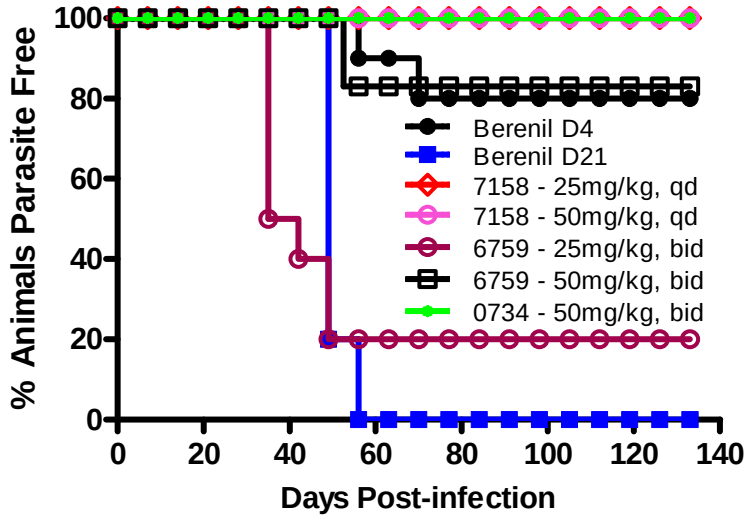
% Animals Parasite Free
100
80
60
40
20
0
0
20
40
60
80
100
120
140
Days Post-infection
Berenil D4
Berenil D21
7158 - 25mg/kg, qd
7158 - 50mg/kg, qd
6759 - 25mg/kg, bid
6759 - 50mg/kg, bid
0734 - 50mg/kg, bid

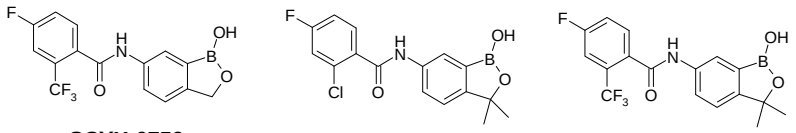

SCYX-6759 SCYX-0734 SCYX-7158

- 3,3-Disubstituted oxaboroles show improved efficacy in a murine model for Stage II Trypanosomiasis.

SCYX-7158 – Optimized Lead Profile

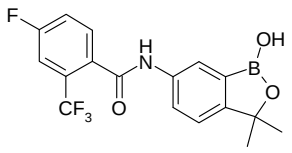

SCYX-7158

T.b.brucei IC_{50} = 556nM
Cytotoxicity (L929) > 25 μM
LogD = 3.51
Solubility (pH 7.4) = 25 μM
S9 $t_{1/2}$ > 350 min (mouse, rat, human)
P_{app} = 414.8 nm/s (AQ = 0.03)
Oral Bioavailablity (mouse) = 57%

Summary

- Oxaboroles have been identified as a promising lead series for the treatment of HAT; demonstrating high *in vitro* potency vs. *T. b. brucei* and good PK and physiochemical properties.
- Lead optimization of the 6-N benzamide region has afforded compounds that are orally active in murine models of both acute (stage I) and chronic (stage II) trypanosomiasis.
- Functionalization of the C-3 position on the oxaborole ring has significantly improved overall PK properties, particularly by increasing CNS disposition and retention.
- The optimized lead compound (SCYX-7158) is currently demonstrating robust *in vivo* activity against stage II trypanosomiasis at clinically relevant dosing levels in ongoing studies.
- Further biological, toxicological and pharmacokinetic profiling of SCYX-7158 is also in progress.