Weight gain in children initiating ART is not increased by nutritional supplements

Poster 718

www.arrowtrial.org
Email: arrow@ctu.mrc.ac.uk

Mutsa Bwakura-Dangarembizi[1], Annette Babirye[2], Adrian Cook[3], Christine Karungi[4], Philip Kasirye[2], Fabbie Mapinge[1], Moses Musinguzi[5], Patricia Nahirya-Ntege[5], Josephine Najjuko[4], Andrew Prendergast[3] on behalf of the ARROW trial team

1 University of Zimbabwe College of Health Sciences, Harare, Zimbabwe. 2 Mulago Hospital, Kampala, Uganda. 3 Medical Research Council Clinical Trials Unit, London, UK.
4 Joint Clinical Research Centre, Kampala, Uganda. 5 Medical Research Council/Uganda Virus Research Institute, Entebbe, Uganda

ABSTRACT

Background Malnutrition among HIV-infected children in resource-limited settings is common, but rapid weight gain usually follows ART initiation. The effect of ready-to-use therapeutic foods (RUTF) in this context is unclear. The nutritional status of a child is likely to influence both the use and effect of RUTF, so causal models are required to establish benefit.

Methods ARROW is an ongoing randomised trial of monitoring and treatment strategies in ART-naïve children in Uganda/Zimbabwe. Enrolment was from May 2007 to November 2008; nutritional supplement data was collected from December 2007. Three of four centres gave Plumpy'nut, a peanut-based RUTF, using criteria that included weight-for-height, mid-upper arm circumference, clinical status and knowledge of food insecurity. Marginal structural models with stabilized time-dependent inverse probability treatment weights were used to estimate the causal effect of Plumpy'nut on weight-for-age Z-scores (WAZ) over the first 48 weeks on ART among children aged <9 years who were enrolled after December 2007, stratifying by centre to compute weights.

Results 387 children aged <9years at ART initiation were enrolled from the 3 centres after Dec 2007; 207 (53%) were female, median(IQR) age was 3(1,6) years, weight-for-age Z-score -1.8(-2.9,-1.0), height-for-age Z-score -2.4(-3.4,-1.3) and weight-for-height Z-score -0.6(-1.4,0.2). Plumpy'nut was given to 66 (17%) children for median(IQR) 17(6,30) weeks, starting 2(2,8) weeks after enrolment. Time-dependent predictors of Plumpy'nut use were age, WAZ, weight <80% expected, mid-upper arm circumference and previous hospital admission. Children given Plumpy'nut had lower WAZ at baseline (-4.0(-4.7,-3.0) vs -1.5(-2.2,-0.8)). In unadjusted analysis, median improvement in WAZ at 48 weeks was greater among those receiving Plumpy'nut (+2.0 vs +0.5); however this reflected greater weight gain in children starting from very low WAZ regardless of Plumpy'nut use. In adjusted analysis, the causal effect of Plumpy'nut was small and not statistically significant (+0.02 per month 95%CI (-0.03,0.08); p=0.45).

Conclusion Malnourished HIV-infected children initiating ART plus RUTF generally experience rapid weight gain. However, a marginal structural model applied to observational data with time-varying confounders suggests that the weight gain was not attributable to the use of RUTF. Routine use of RUTF for stable, malnourished children entering ART programs may therefore not be clinically beneficial.

METHODS

- HIV-infected ART-naïve children meeting WHO criteria for ART in Uganda/Zimbabwe were enrolled into the ARROW trial during 2007-2008 and started combination ART
- 4-weekly follow-up included measurement of weight and height
- Malnutrition during follow-up was treated in an outpatient setting using the peanut-based RUTF Plumpy'nut
- Weight gain in the first 48 weeks was analysed, corresponding to the period of most rapid growth and highest RUTF use
- In children aged 3 months to 17 years, weight and height were analysed as weight-for-age z-score (WAZ) and height-for-age z-score (HAZ), using WHO 2007 reference data
- Children with unstable severe acute malnutrition (SAM) were hospitalized for therapeutic feeding prior to enrolment into ARROW

RESULTS

1206 children enrolled to ARROW

excluded

- Age 10 years before 48 weeks follow-up - normalised data unavailable, n=327
- One trial site provided nutritional support externally - incomplete supplement data, n=267
- Children enrolled before 1 Dec 2007: RUTF use not recorded before this date, n=225

387 children eligible for analysis of RUTF and weight gain

Baseline participant characteristics

			N=387
Sex	**boys**	**n (%)**	180 (47%)
Age	**years**	**median (IQR)**	3.3 (1.6,6.1)
WHO stage	**1**	**n (%)**	10 (3%)
	2		141 (36%)
	3		169 (44%)
	4		67 (17%)
CD4%	**< 5 yrs**	**median (IQR)**	15% (11%,20%)
CD4	**≥5 yrs**		280 (122,503)
Weight-for-age	**z-score**	**median (IQR)**	-1.9 (-2.8,-1.0)
Height-for-age			-2.4 (-3.4,-1.3)

RUTF and WEIGHT GAIN – unadjusted analysis

- 66 children (17%) given RUTF during first 48 weeks of ART
 - started median 2 weeks after enrolment (IQR 2,8)
 - continued for median 17 weeks (IQR 6,30)
- Baseline WAZ lower in children given RUTF
 -4.0 vs -1.5 in users and non-users, respectively
- Greater WAZ increase in children given RUTF
 +2.0 v +0.5 in users and non-users, respectively

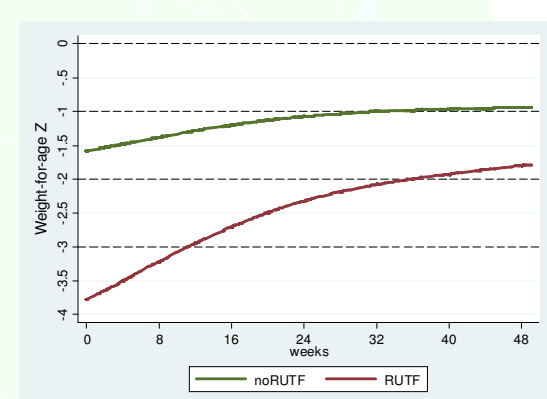
Weight-for-age Z
0
-.5
-1
-1.5
-2
-2.5
-3
-3.5
-4
0
8
16
24
32
40
48
weeks
noRUTF
RUTF

MODELLING

A marginal structural model was used since the use of RUTF was subject to time-varying confounders.

Predictors in treatment model were weight-for-age at previous visit, WHO stage 4 HIV disease, hospitalisation since previous visit, age, month from ART initiation, and a composite variable of either mid-upper-arm-circumference or weight-for-height indicating the need for therapeutic feeding.

Stabilized treatment weights were applied to a repeated measures generalised estimating equation model of weight-for-age z-score with one-level autoregressive correlation.

The cumulative effect of RUTF is estimated as days from start of use, remaining constant after use of RUTF ended. Under this model a child may grow at a faster rate while using RUTF; after its use ends the additional growth is then maintained.

Change over time was modelled using cubic splines as above with resulting effect estimates approximated by piecewise linear slopes changing at 16 weeks.

RUTF and WEIGHT GAIN – adjusted analysis

Mean change in WAZ = +0.02 per month (95%CI -0.03,0.08, p=0.45)

The effect of RUTF on weight gain is small and not statistically significant

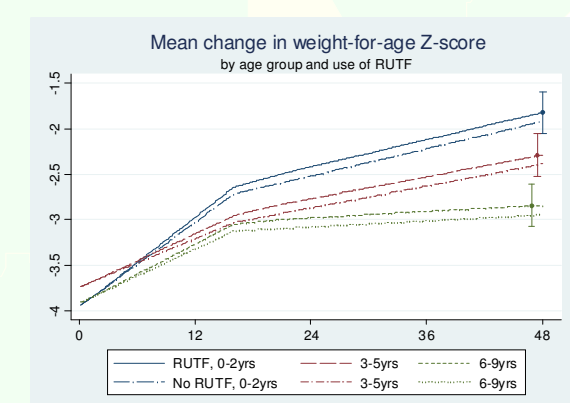
Mean change in weight-for-age Z-score
by age group and use of RUTF
-1.5
-2
-2.5
-3
-3.5
-4
0
12
24
36
48
RUTF, 0-2yrs
No RUTF, 0-2yrs
3-5yrs
3-5yrs
6-9yrs
6-9yrs

SUMMARY

Children showed excellent growth during their first year on ART. However, causal modelling suggests that RUTF does not significantly increase weight gain in children initiating ART. Children given RUTF did have greater increases in weight-for-age, but marginal structural modelling showed that this association was not causal. Growth reconstitution in HIV-infected children appears to be mediated primarily by ART. The results of this observational analysis should be confirmed with a randomized trial of RUTF. The REALITY trial, starting in 2012, will include ready-to-use supplementary food in a factorial design of interventions to evaluate strategies to reduce early mortality in adults and children starting ART.

We thank all the patients and staff from all the centres participating in the ARROW trial. Joint Clinical Research Centre, Kampala, Uganda: P Mugyenyi, V Musiime, VD Afayo, E Bagurukira, J Bwomezi, J Byaruhanga, P Erimu, C Karungi, H Kizito, M Mutumba, WS Namala, J Namusanje, R Nandugwa, TK Najjuko, E Natukunda, M Ndigendawani, SO Nsiyona, F Odongo, K Robinah, M Ssenyonga, D Sseremba, J Tezikyabbiri, CS Tumusiime; **MRC/UVRI Uganda Research Unit on AIDS, Entebbe, Uganda**: P Munderi, P Nahirya-Ntege, M Aber, FN Kaggwa, P Kaleebu,R Katuramu, JH Kyalimpa, J Lutaakome, L Matama, M Musinguzi, G Nabulime, A Ruberantwari, R Sebukyu, IM Ssekamatte, G Tushabe, D Wangi. **Baylor-Uganda, Paediatric Infectious Disease Centre, Mulago Hospital, Uganda**: A Kekitiinwa, P Musoke, S Bakeera-Kitaka, R Namuddu, P Kasirye, JK Balungi, A Babirye, J Asello, S Nakalanzi, NC Ssemambo, J Nakafeero, JN Kairu, EK George, G Musoba, J Ssanyu, S Ssenyonjo. **University of Zimbabwe, Harare, Zimbabwe**: KJ Nathoo, MF Bwakura-Dangarembizi, F Mapinge, T Mhute, T Vhembo, R Mandidewa, D Nyoni, C Katanda, GC Tinago, J Bhiri, D Muchabaiwa, S Mudzingwa, MM Chipiti, M Phiri, J Steamer, CC Marozva, SJ Maturure, L Matanhike, S Tsikirayi, L Munetsi. **Medical Research Council Clinical Trials Unit, London, UK:** DM Gibb, MJ Thomason, AD Cook, A Prendergast, AA Ferrier, B Naidoo, MJ Spyer, AS Walker, LK Kendall. **Independent DART Trial Monitors:** R Nanfuka. **Trial Steering Committee**: I Weller (Chair), E Luyirika, H Lyall, E Malianga, C Mwansambo, M Nyathi, A Wapakhabulo, DM Gibb, A Kekitiinwa, P Mugyenyi, P Munderi, KJ Nathoo: Observers S Kinn, M MacNeil, M Roberts, W Snowden. **Data and Safety Monitoring Committee**: A Breckenridge (Chair), C Hill, J Matenga, A Pozniak, J Tumwine. **Endpoint Review Committee**: G Tudor-Williams (Chair), H Barigye, HA Mujuru, G Ndeezi. **Funding**: ARROW is funded by the **UK Medical Research Council** and the **UK Department for International Development** (DfID). Drugs are provided by **GlaxoSmithKline.**